Next-generation core inhibitors demonstrate increased potency and distinct biochemical properties compared to first-generation core inhibitors

131

Nuruddin Unchwaniwala[1], Steven P Dunkelbarger[2], Adam Zlotnick[2,3], Karolyn Pionek[4], Daniel D Loeb[4], William Delaney[1], Kathryn M Kitrinos[1]

[1]Assembly Biosciences, Inc., South San Francisco, CA, USA; [2]Door Pharmaceuticals, LLC., Bloomington, IN, USA; [3]Indiana University, Bloomington, IN, USA; [4]University of Wisconsin-Madison, Madison, WI, USA

BACKGROUND

- Core inhibitors are a novel class of antivirals with the potential to improve cure rates in patients with chronic hepatitis B virus infection (cHBV). These agents:
 - Inhibit multiple steps in the viral replication cycle, including pregenomic (pg)RNA encapsidation, which blocks the assembly and release of new viral particles containing pgRNA or HBV DNA and disruption of incoming capsids, which blocks the establishment of de novo covalently closed circular (ccc)DNA during infection (**Figure 1**)[1]
 - Demonstrated potent antiviral activity in Phase 1 studies[2,3] and enhanced antiviral activity when combined with nucleos(t)ide reverse transcriptase inhibitors (NrtIs) in Phase 2 studies[4,5]
- Next-generation core inhibitors, such as ABI-H3733 (3733) and ABI-4334 (4334), have the potential to provide deeper virologic suppression by providing greater inhibition in the formation of cccDNA compared to first-generation core inhibitors[6,7]

Figure 1. Core Inhibitor Mechanisms of Action

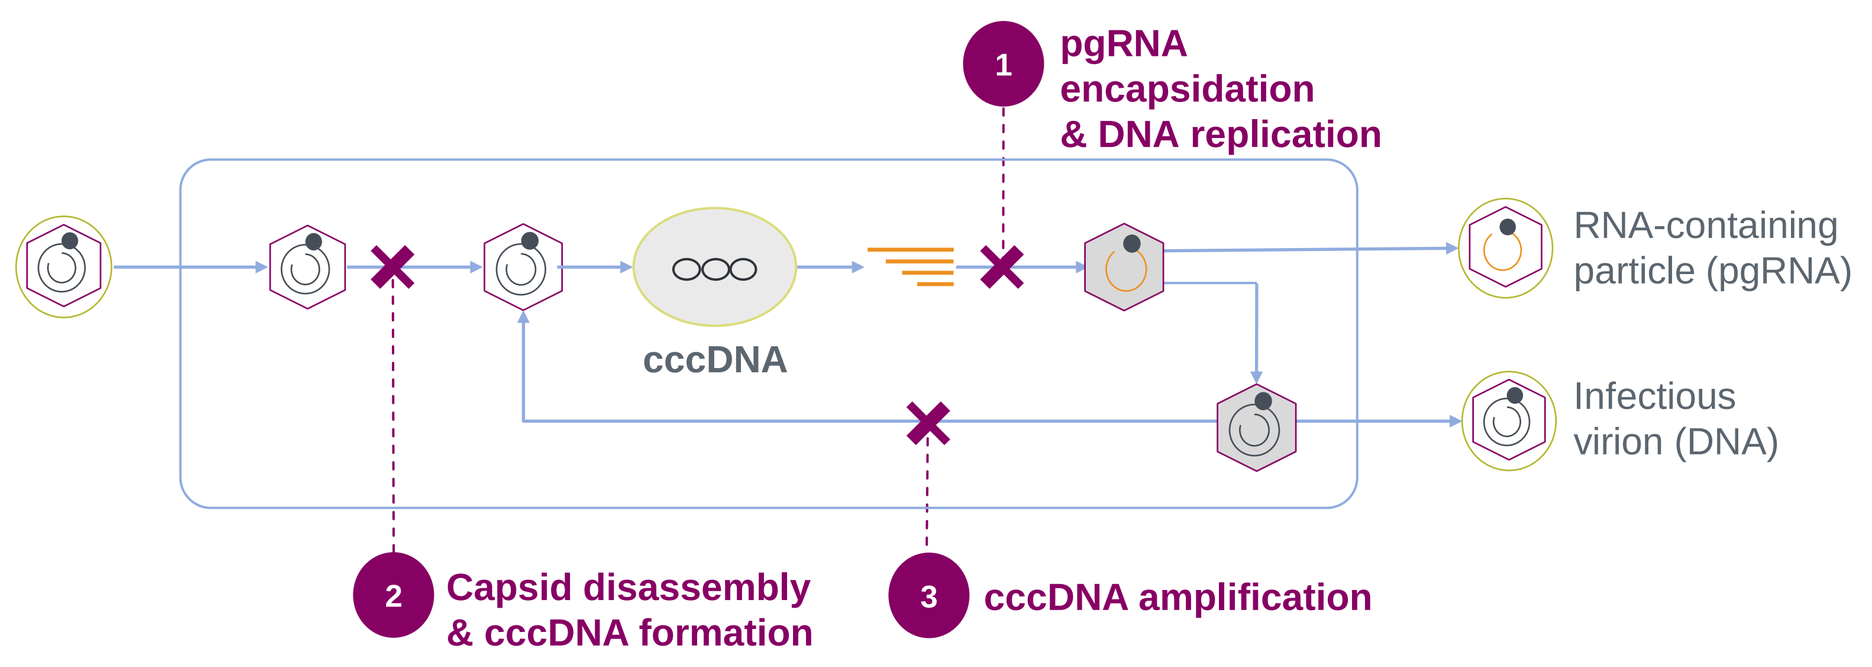
1 pgRNA encapsidation & DNA replication
cccDNA
RNA-containing particle (pgRNA)
Infectious virion (DNA)
2 Capsid disassembly & cccDNA formation
3 cccDNA amplification

cccDNA, covalently closed circular DNA; pgRNA, pregenomic RNA.

OBJECTIVE OF THIS ANALYSIS

- To characterize the potency and biochemical properties of 3733 and 4334, two next-generation, novel, investigational core inhibitors with high potency against pgRNA encapsidation and cccDNA formation

METHODS

- **Primary human hepatocyte (PHH) assays**
 - **Intracellular replication and hepatitis B e antigen (HBeAg) secretion:** PHHs were infected with HBV at multiplicity of infection (MOI) of 300 viral genome equivalents (vge)/cell for 3 hours, followed by drug treatment. Cells were retreated on Day 4, and cultures were harvested on Day 8. Intracellular HBV DNA was measured using a branched DNA assay and secreted HBeAg was measured via an enzyme-linked immunosorbent assay
 - **Direct cccDNA quantification:** PHHs were infected with HBV at MOI of 500 vge/cell for 3 hours, followed by drug treatment. At Day 4, cells were harvested, and total DNA was extracted using a modified Hirt DNA-extraction procedure. Total DNA was treated with T5 exonuclease to digest linear and nicked double-stranded DNA, followed by EcoRI digestion to linearize cccDNA. Extracted cccDNA was analyzed by Southern blot probing against HBV DNA using branched DNA signal amplification and detection
- **HepAD38 assays**
 - **Capsid analysis:** Induced and treated HepAD38 cells were lysed at Day 4 with 1% NP-40 lysis buffer, and cytoplasmic fractions were electrophoresed on native agarose gels and blotted onto a polyvinylidene difluoride membrane for core immunostaining using in-house rabbit polyclonal core antibody and onto nylon membrane for probing against HBV DNA
 - **Velocity sedimentation:** A fraction of the capsid analysis cytoplasmic lysate was loaded onto a step sucrose gradient (15%–60% sucrose) and the gradient was centrifuged for 2.5 hours at 277,000 g at 10°C. Twelve fractions were separated from top to bottom. Each fraction was run on a sodium dodecyl sulfate-polyacrylamide gel, followed by western blot detection of core protein
- **Biochemical studies**
 - **Capsid assembly:** Assembly of purified core protein was initiated by adjusting the reaction conditions to 3.5 μM core, 3 μM core inhibitor, 150 mM sodium chloride (NaCl), and 50 mM hydroxyethyl piperazineethansulfonic acid (HEPES). Negative-stained samples were prepared on grids after 30 minutes or 72 hours of incubation and imaged via transmission electron microscopy
 - **Capsid disruption:** Capsids were assembled from BODIPY-labeled core protein and then diafiltered to remove free subunits. The capsid suspension buffer was adjusted to 50 mM HEPES, 20 mM NaCl, and 300 mM urea to mimic the fragility of relaxed circular DNA-containing capsids. After overnight incubation in the presence of various core inhibitors or 300 mM NaCl (stabilizing control) or 3 M urea (destabilizing control), fluorescence readouts for BODIPY were acquired

RESULTS

Figure 2. 3733 and 4334 Potently Inhibit Intracellular Replication and cccDNA Formation

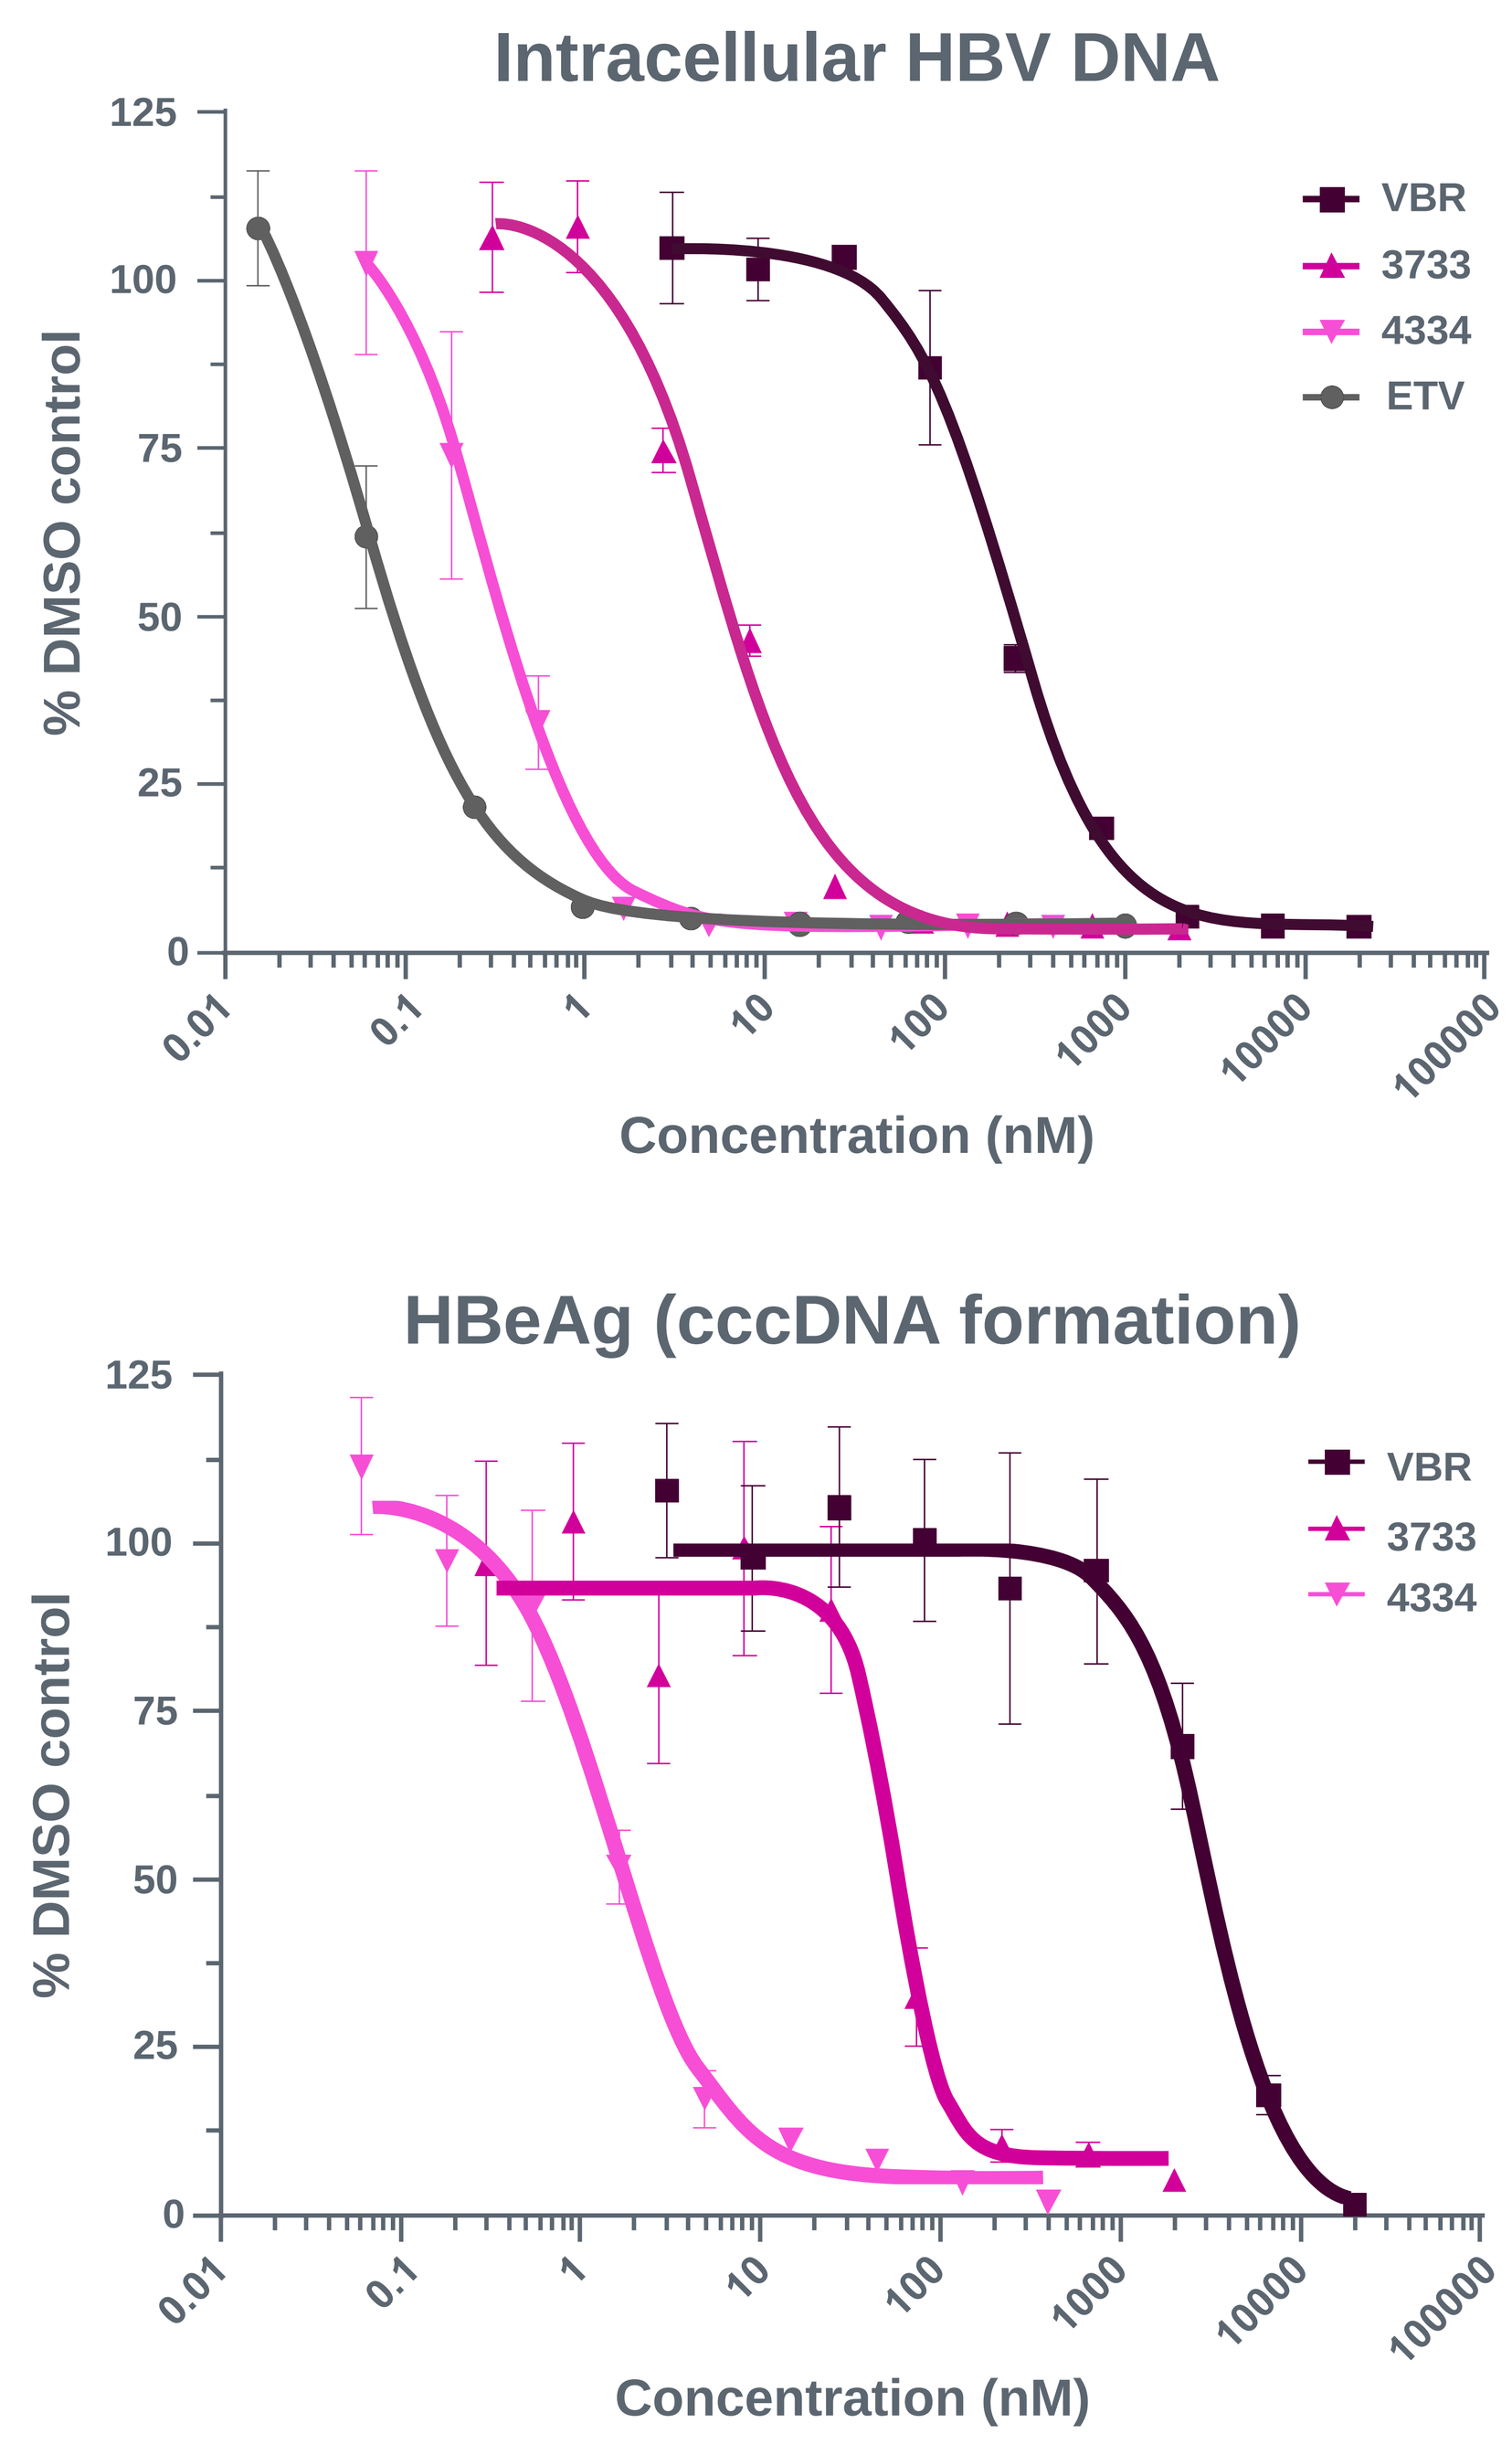
Intracellular HBV DNA
% DMSO control
Concentration (nM)
VBR
3733
4334
ETV
HBeAg (cccDNA formation)
% DMSO control
Concentration (nM)
VBR
3733
4334

	VBR	3733	4334	ETV
PHH DNA replication EC_{50} (nM)	200 ± 14	5.6 ± 0.5	0.3 ± 0.1	0.05 ± 0.01
PHH cccDNA formation EC_{50} (nM)	3298 ± 578	56 ± 8.3	1.5 ± 0.2	>1000
PHH cccDNA formation (direct quantification), EC_{50} (nM)	3443	102	3.1	-

3733, ABI-H3733; 4334, ABI-4334; cccDNA, covalently closed circular DNA; DMSO, dimethyl sulfoxide; EC_{50}, half maximal effective concentration; ETV, entecavir; HBeAg, hepatitis B e antigen; HBV, hepatitis B virus; PHH, primary human hepatocyte; VBR, vebicorvir.

- Compared to the first-generation core inhibitor vebicorvir (VBR), the next-generation core inhibitors 3733 and 4334 more potently inhibited intracellular HBV replication and cccDNA formation in PHH (**Figure 2**)

Figure 3. Core Inhibitors Have a Biphasic Effect on HBV Capsids

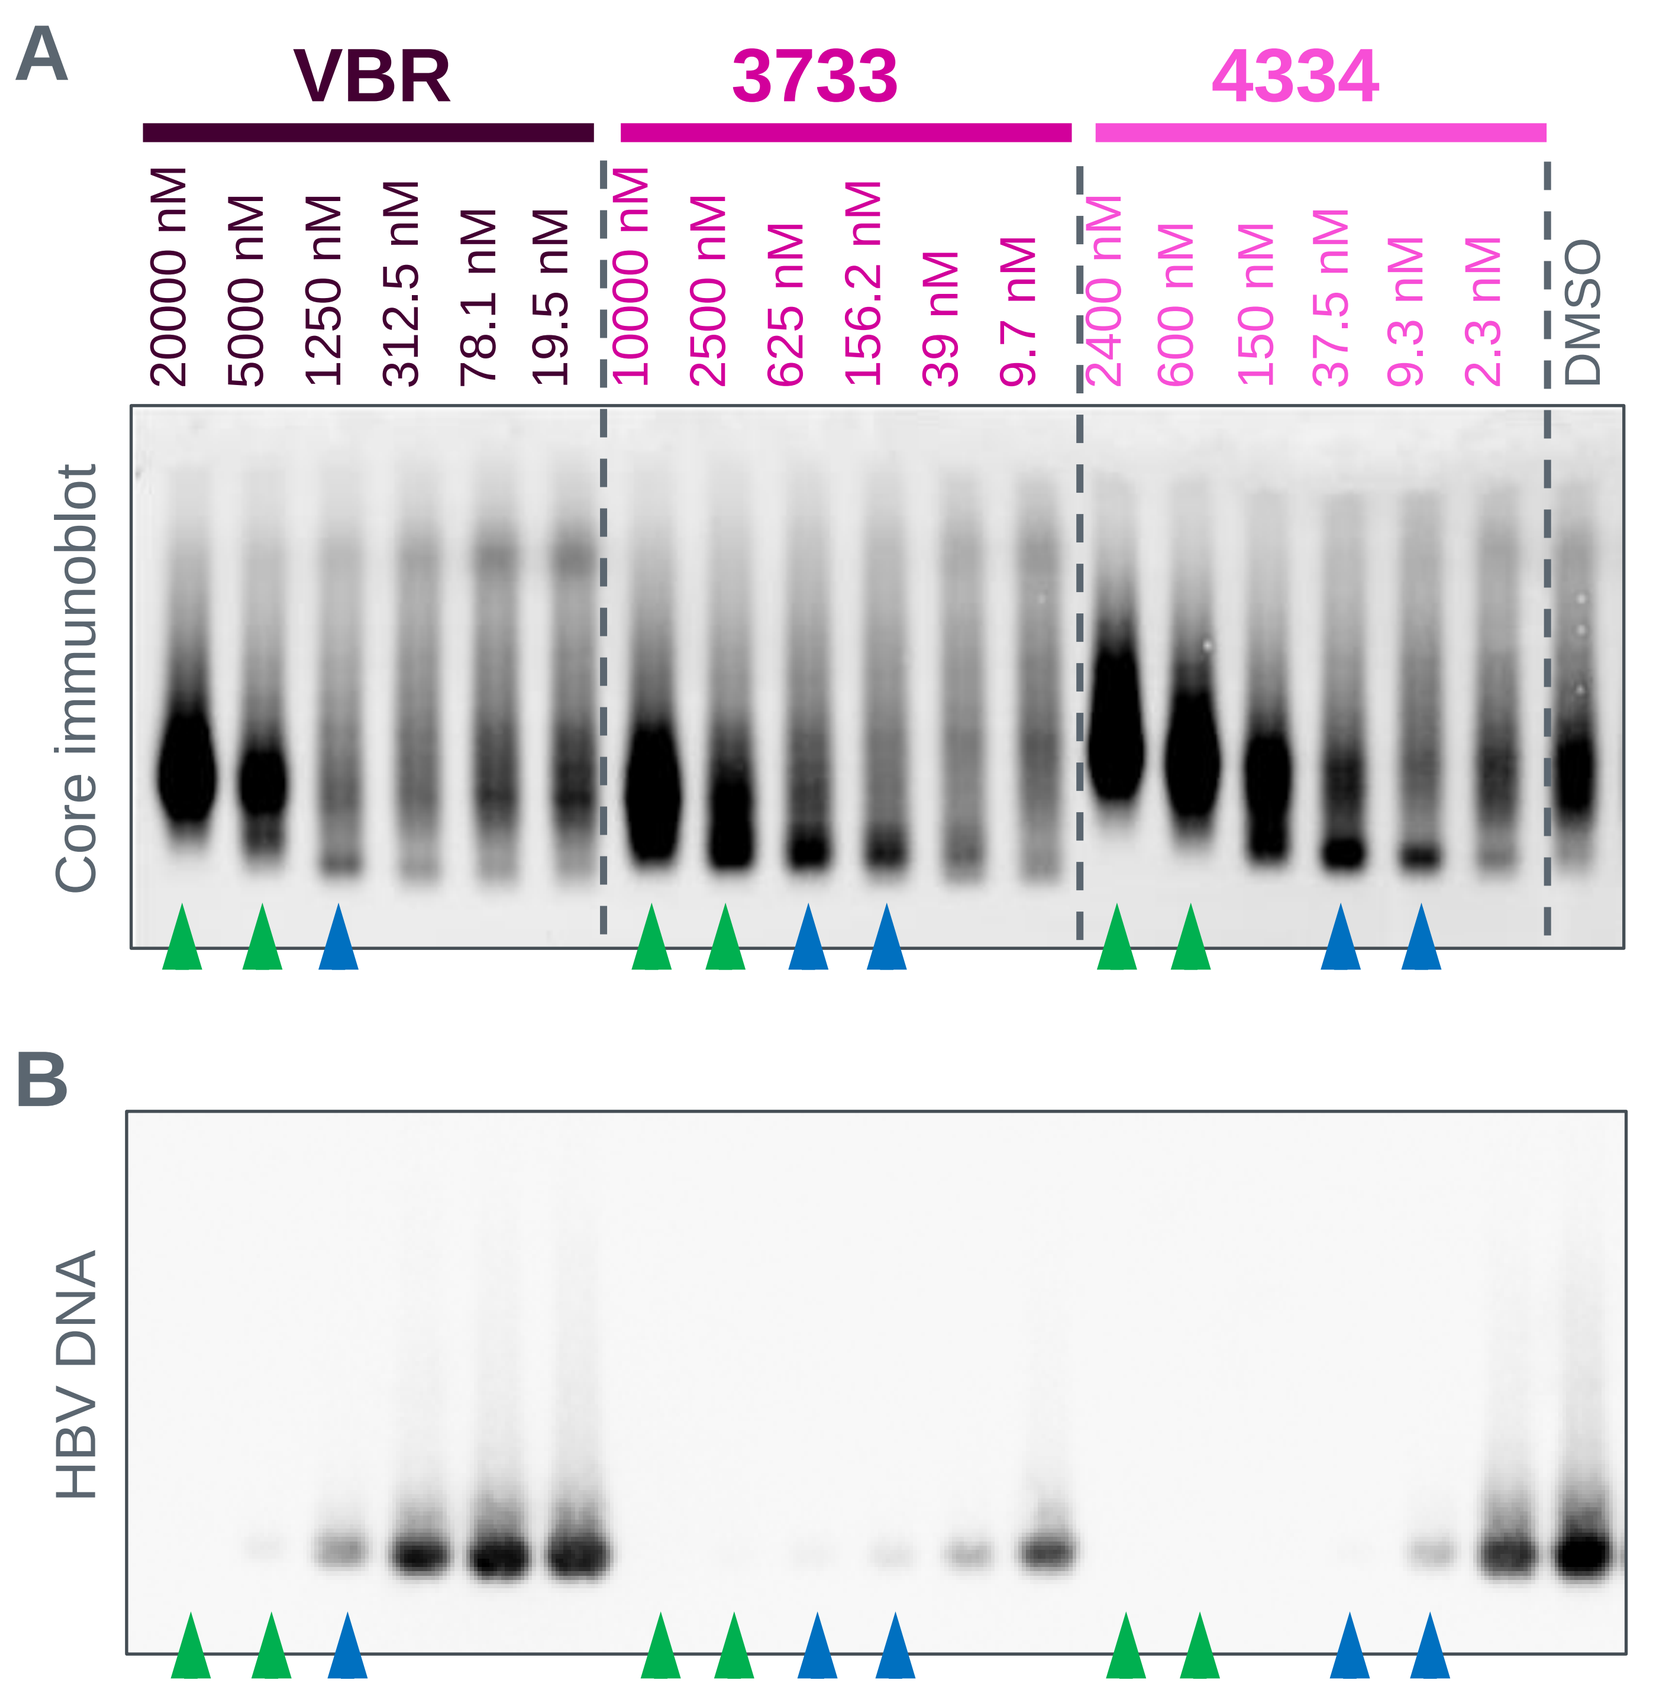
A
VBR
3733
4334
20000 nM
5000 nM
1250 nM
312.5 nM
78.1 nM
19.5 nM
10000 nM
2500 nM
625 nM
156.2 nM
39 nM
9.7 nM
2400 nM
600 nM
150 nM
37.5 nM
9.3 nM
2.3 nM
DMSO
Core immunoblot
B
HBV DNA

3733, ABI-H3733; 4334, ABI-4334; DMSO, dimethyl sulfoxide; HBV, hepatitis B virus; VBR, vebicorvir.

- At lower concentrations of core inhibitors (blue arrowheads), there was an increase in capsids relative to dimethyl sulfoxide (DMSO; **Figure 3A**), with capsids predominantly devoid of HBV DNA (**Figure 3B**). At higher concentrations of core inhibitors (green arrowheads), altered capsid mobility was observed, with unique mobility for each core inhibitor
- The concentrations at which altered capsid mobilities were observed for 3733 (2500 nM) and 4334 (600 nM) are predicted to be clinically achievable,[8,9] while the VBR concentration (5000 nM) is almost 2x higher than the minimum plasma concentration observed in patients with cHBV[2]

Figure 4. All Core Inhibitors Accelerate Assembly of Capsids Devoid of HBV DNA

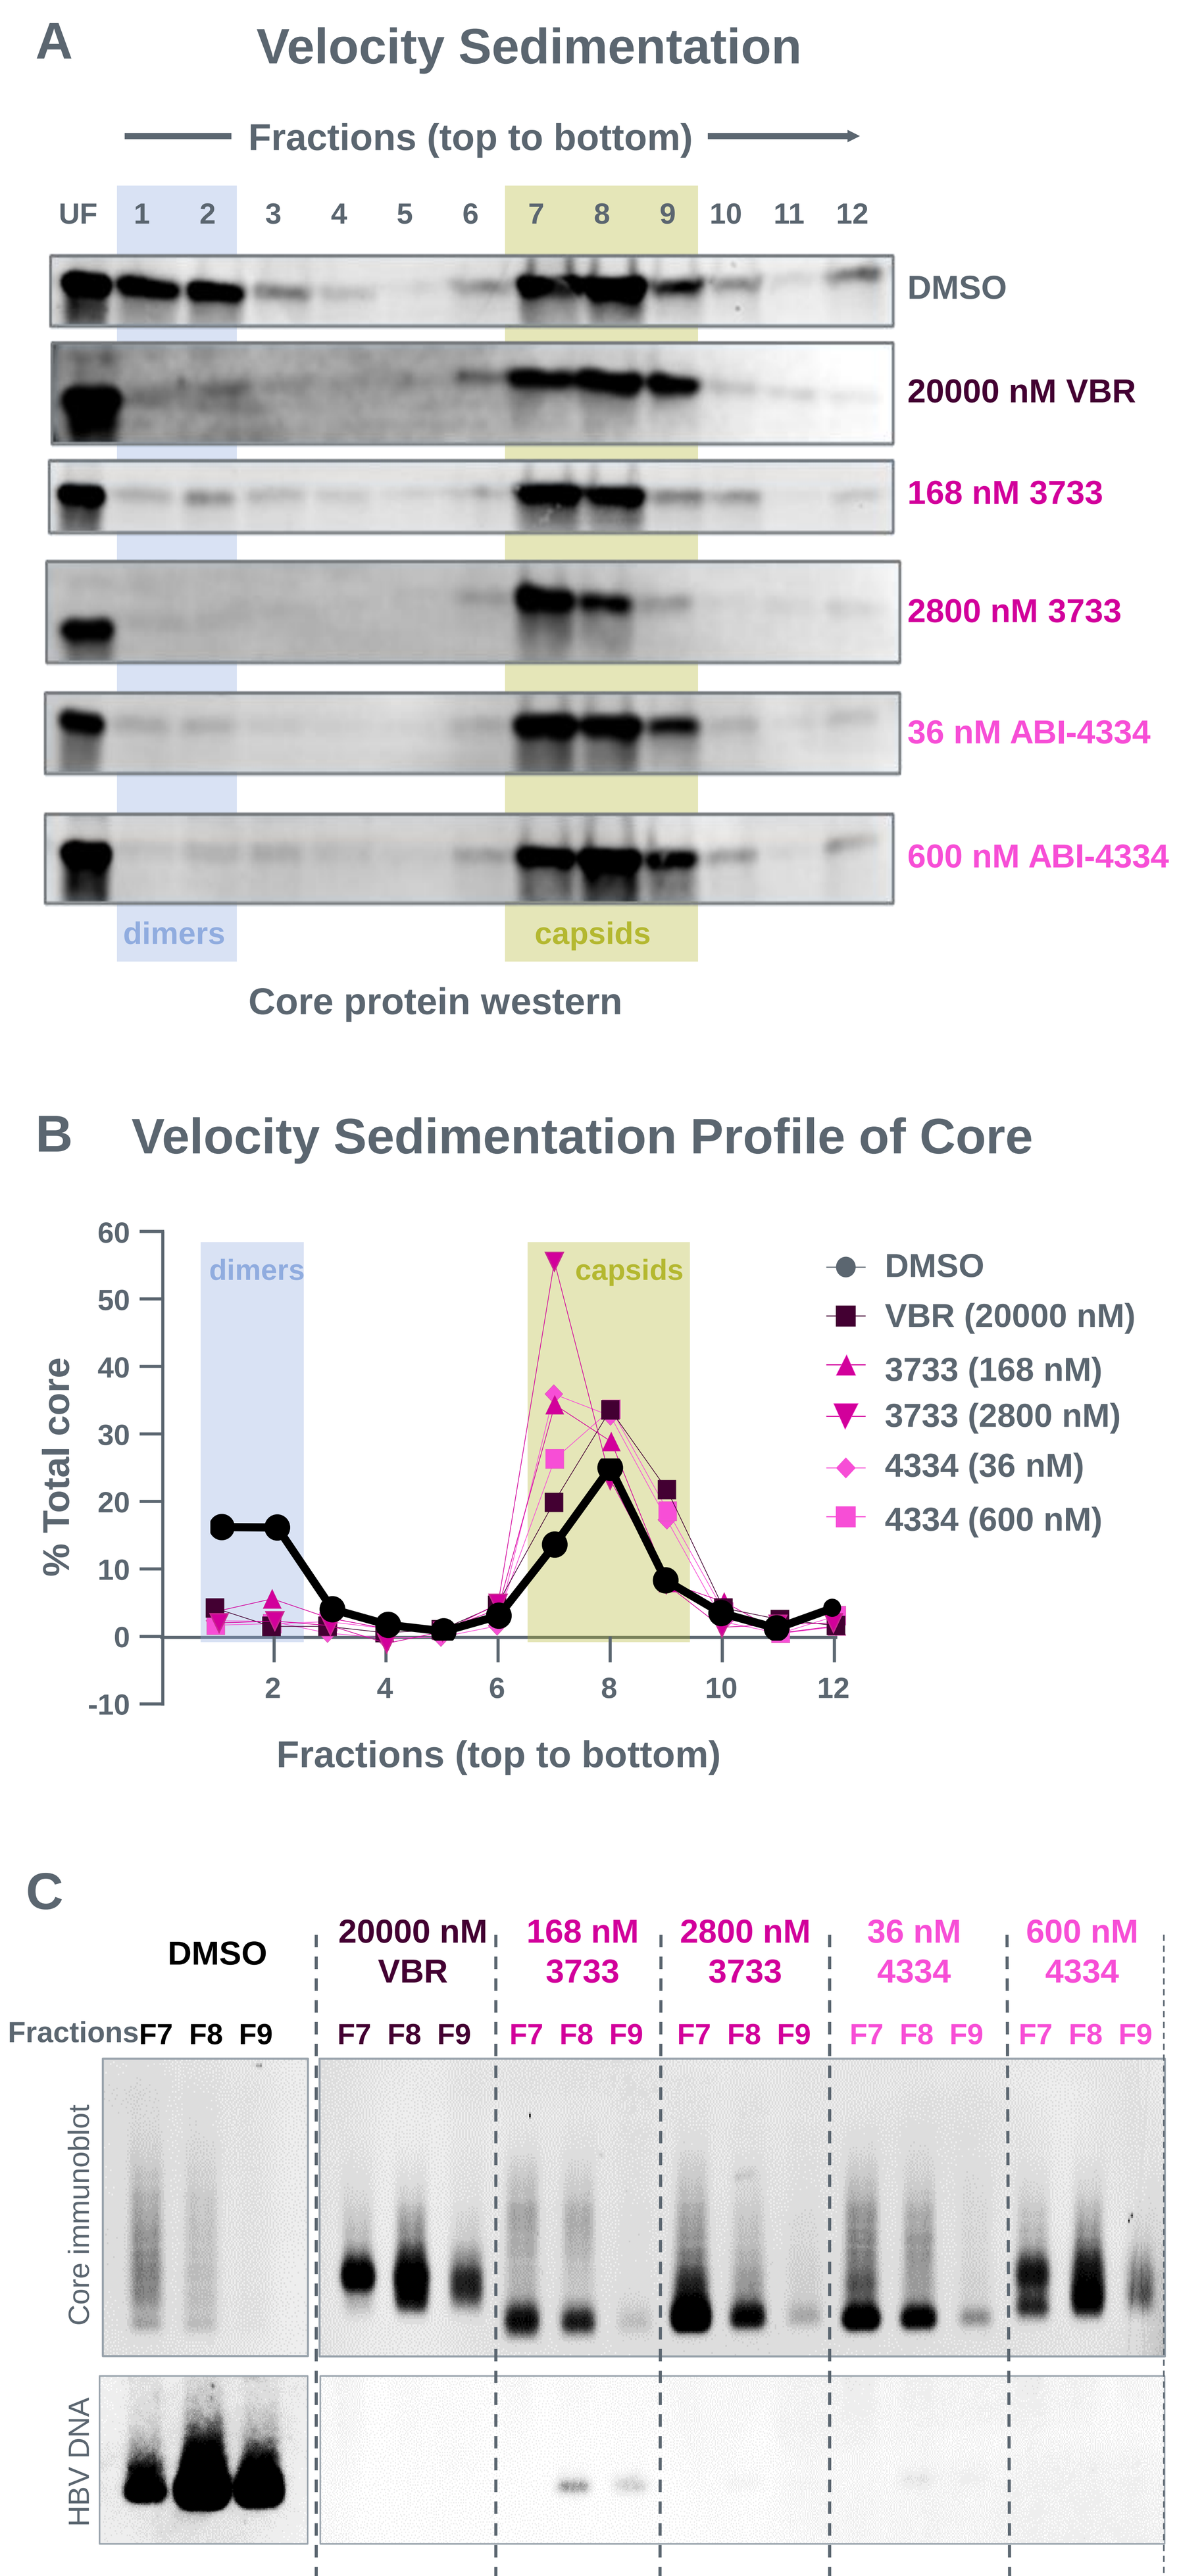
A
Velocity Sedimentation
Fractions (top to bottom)
UF 1 2 3 4 5 6 7 8 9 10 11 12
DMSO
20000 nM VBR
168 nM 3733
2800 nM 3733
36 nM ABI-4334
600 nM ABI-4334
dimers
capsids
Core protein western
B
Velocity Sedimentation Profile of Core
% Total core
Fractions (top to bottom)
dimers
capsids
DMSO
VBR (20000 nM)
3733 (168 nM)
3733 (2800 nM)
4334 (36 nM)
4334 (600 nM)
C
DMSO
20000 nM VBR
168 nM 3733
2800 nM 3733
36 nM 4334
600 nM 4334
Fractions F7 F8 F9
Core immunoblot
HBV DNA

3733, ABI-H3733; 4334, ABI-4334; DMSO, dimethyl sulfoxide; F, fraction; HBV hepatitis B virus; UF, unfractionated; VBR, vebicorvir.

- All core inhibitors accelerated dimer-dimer interactions to predominantly form capsid-like particles (CLPs) as compared to DMSO treated cells (**Figure 4A** and **4B**). Notably, 3733 treated cells had an increased percentage of capsids in fraction 7 compared to VBR and 4334
- Capsid fractions from velocity sedimentation that retained gel migration properties (**Figure 4C**) were identical to the pattern observed with cell lysates (**Figure 3A**), indicating that subtle structural differences in CLPs likely account for the striking gel migration pattern differences

Figure 5. Greater Changes in Capsid Structure Observed with 3733- and 4334-Treated Core Protein

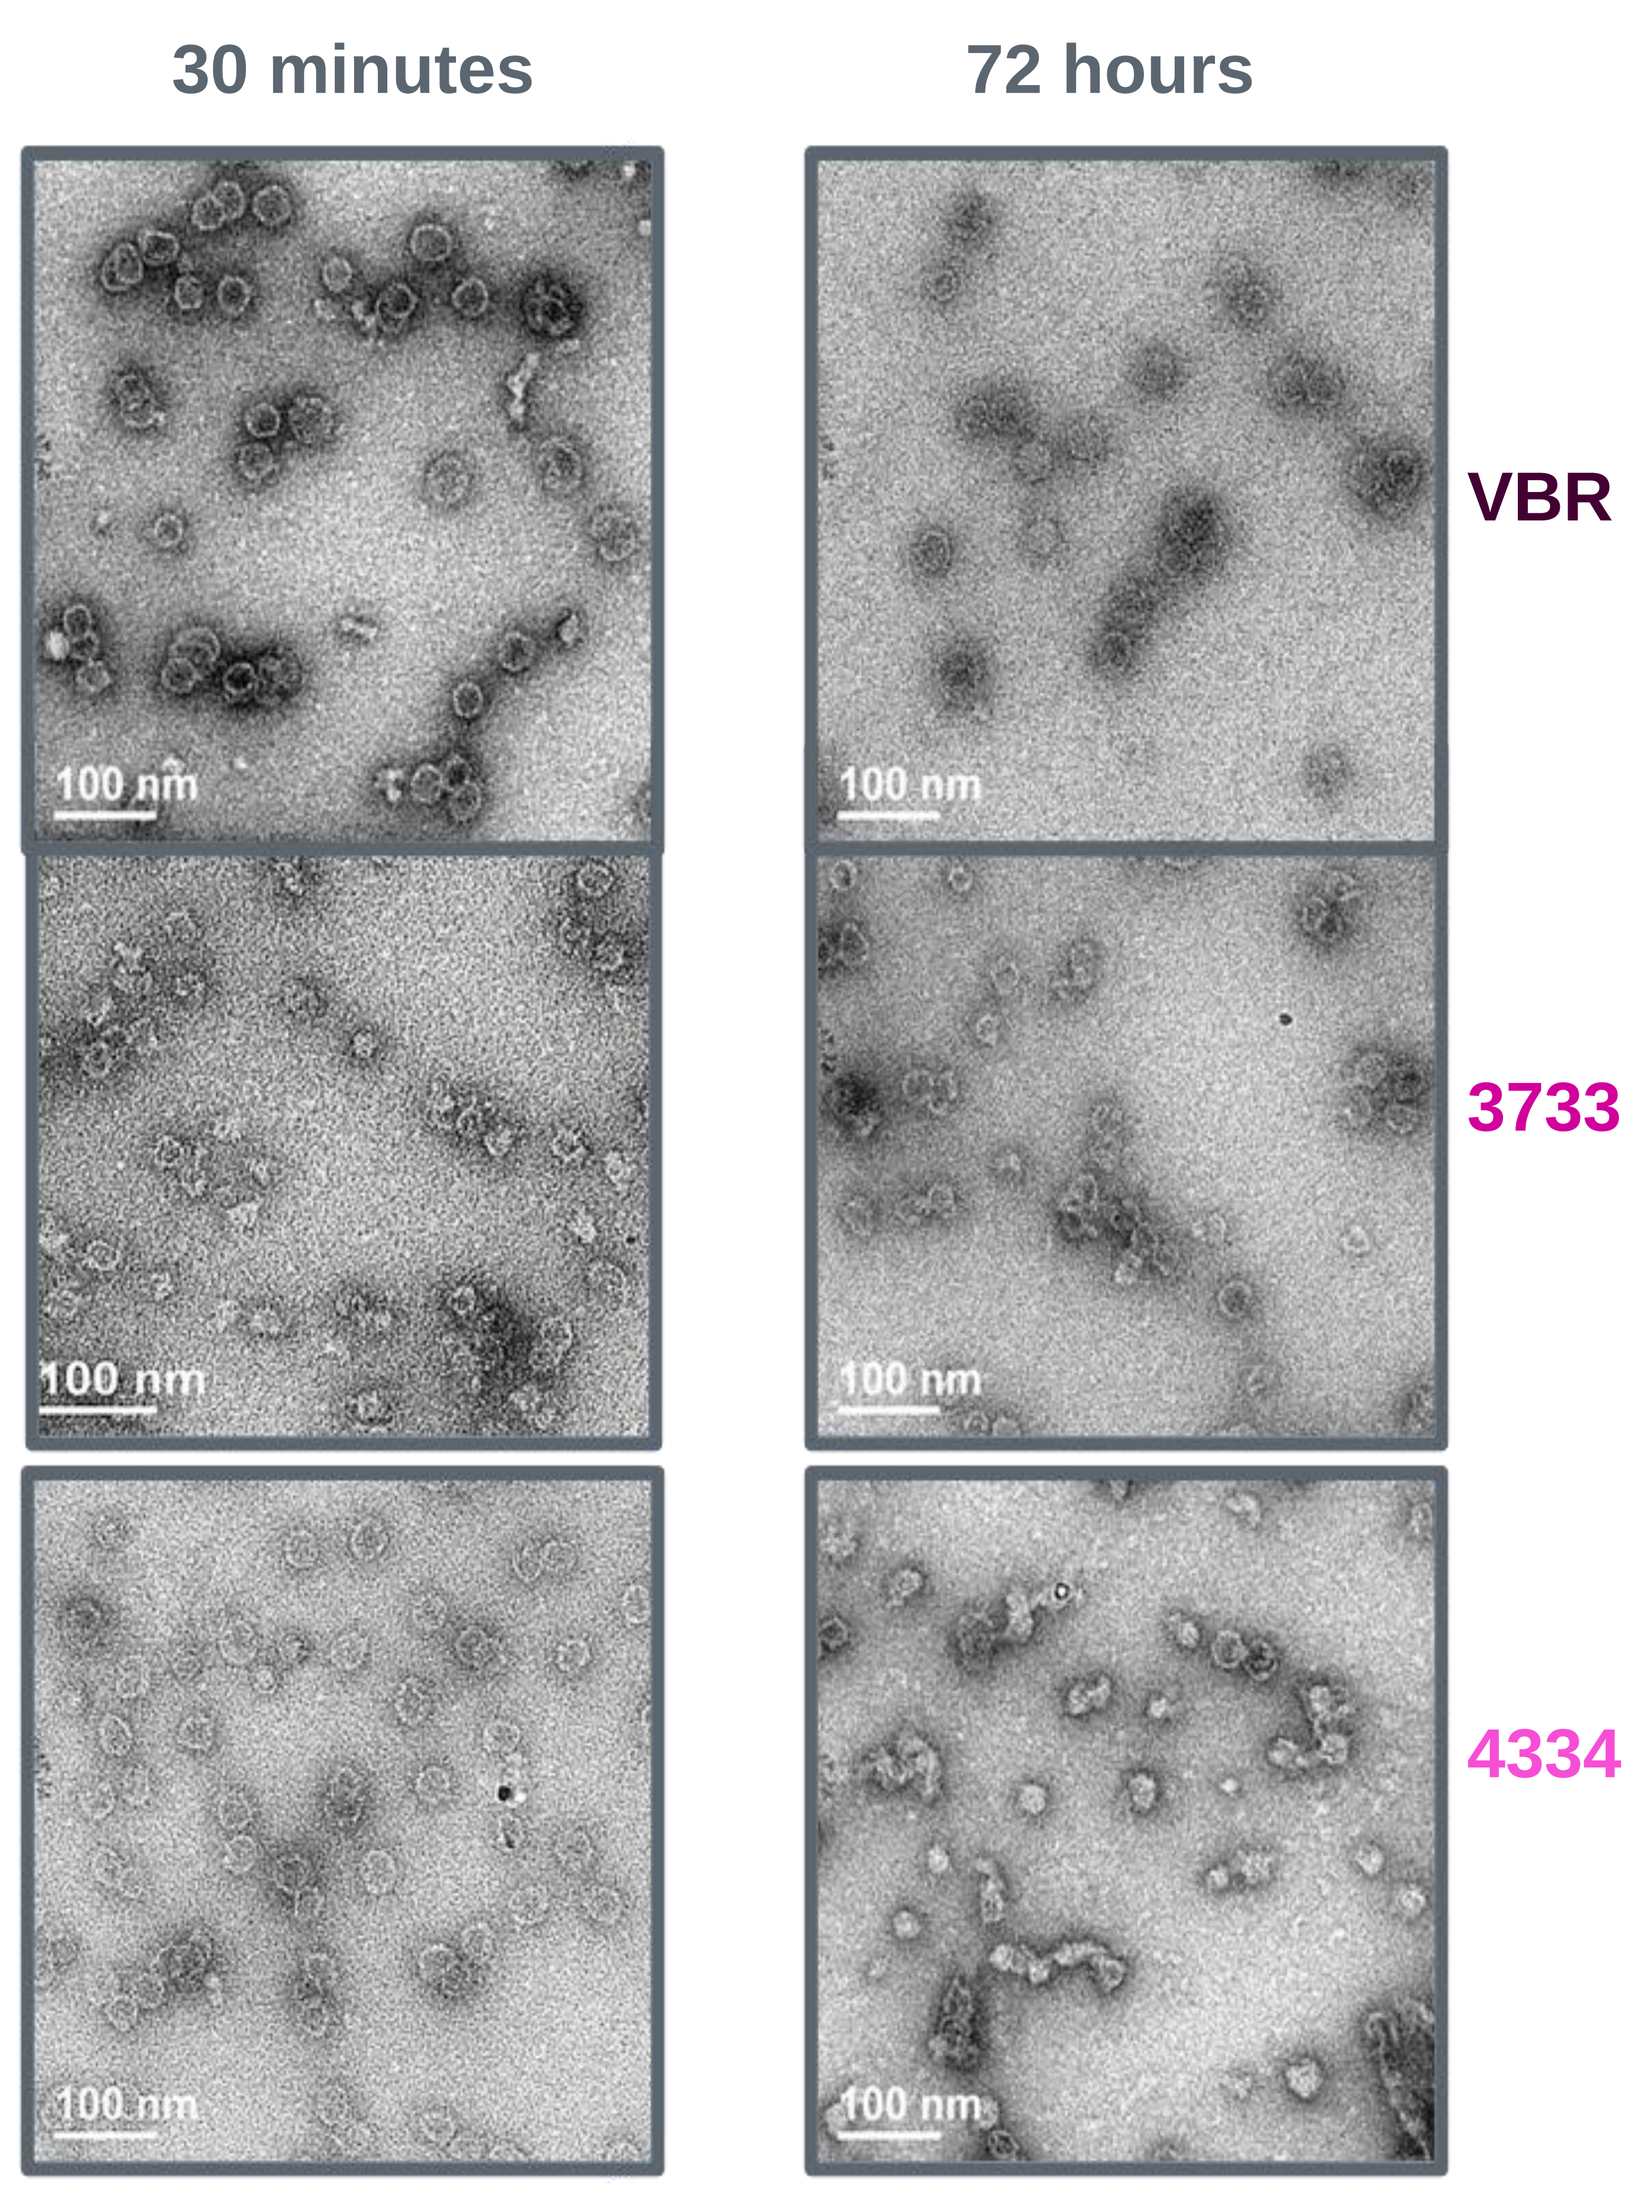
30 minutes
72 hours
VBR
3733
4334
100 nm

3733, ABI-H3733; 4334, ABI-4334; VBR, vebicorvir.

- After short-term incubation of core proteins with 3733 and 4334, CLPs with an increased angularity were observed (**Figure 5**). After longer-term incubations, further capsid rearrangements resulting in condensed cores were observed
- Short- and longer-term incubation of core protein with VBR resulted in both normal and aberrant capsids

Figure 6. 3733 and 4334 Reorganize Capsids Without Releasing Core Subunits

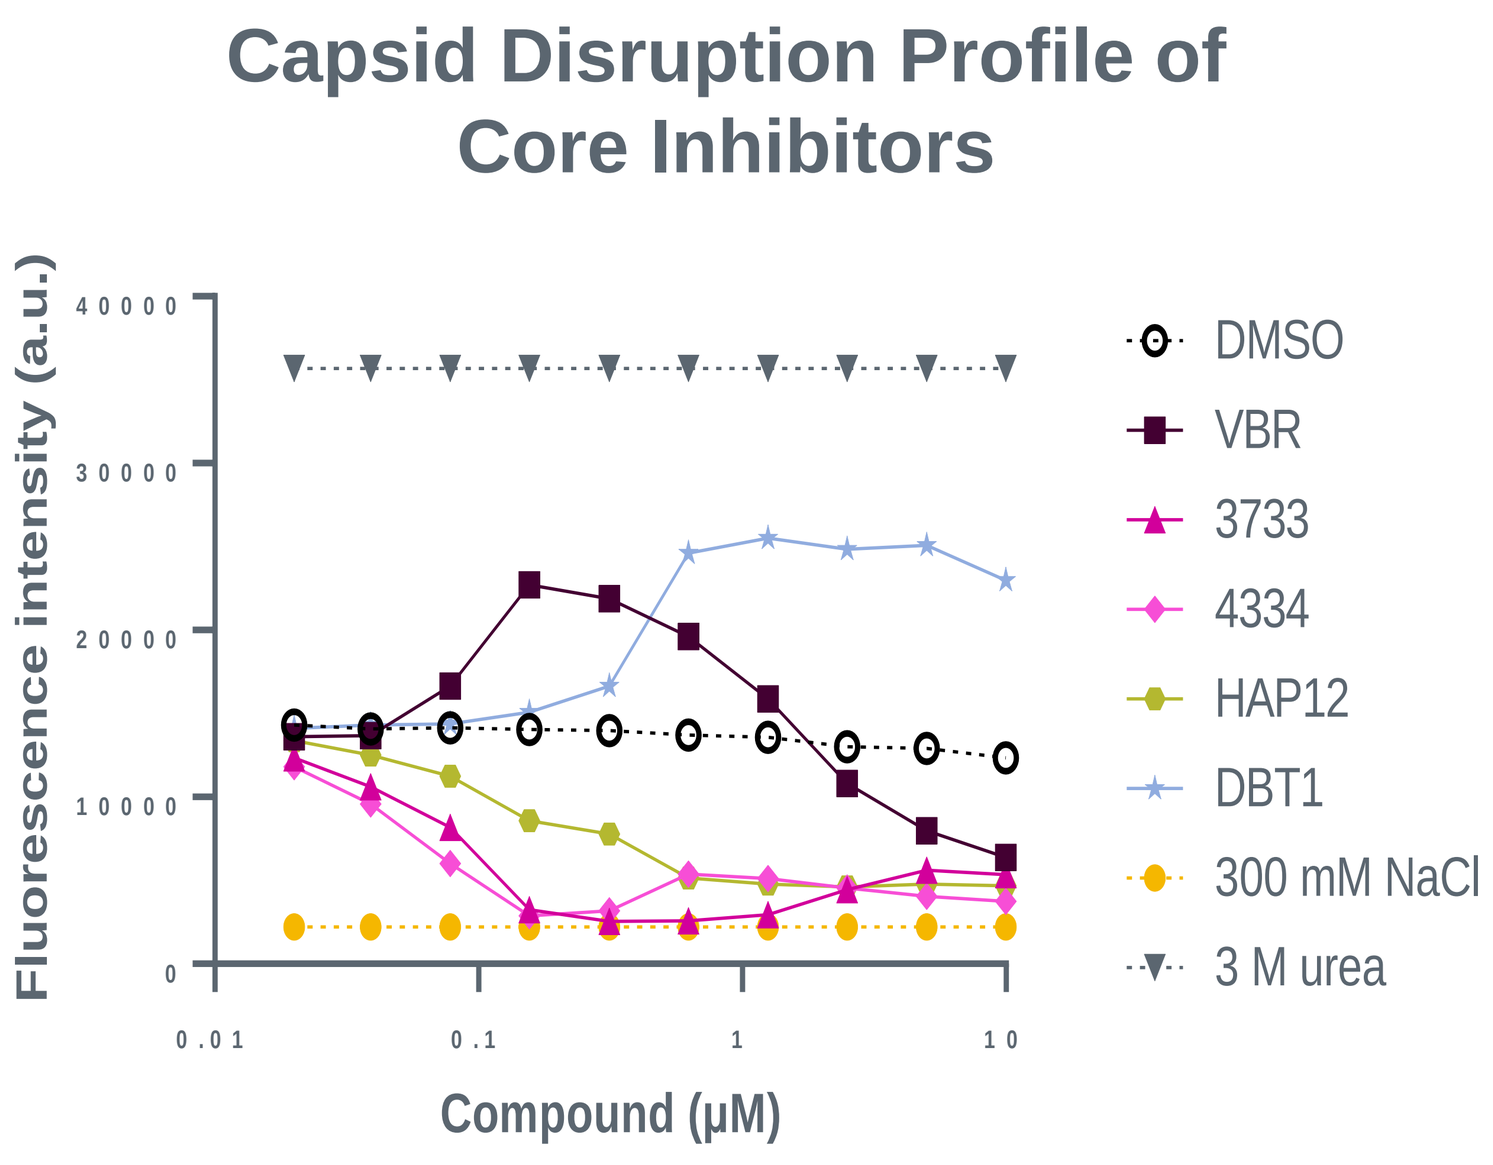
Capsid Disruption Profile of Core Inhibitors
Fluorescence intensity (a.u.)
Compound (μM)
DMSO
VBR
3733
4334
HAP12
DBT1
300 mM NaCl
3 M urea

3733, ABI-H3733; 4334, ABI-4334; a.u., arbitrary units; DBT1, dibenzothiazepine-2-one; DMSO, dimethyl sulfoxide; HAP12, heteroarylpyrimidine 12; NaCl, sodium chloride; VBR, vebicorvir.

- All concentrations of 3733 and 4334 resulted in the condensing of viral capsids; in contrast, VBR triggered the release of core subunits at most concentrations tested (**Figure 6**)

CONCLUSIONS

- 3733 and 4334, two next-generation investigational core inhibitors, have markedly improved potency against both pgRNA encapsidation and cccDNA formation compared to the first-generation investigational core inhibitor VBR
- 3733 and 4334 have distinct biochemical profiles compared to VBR
- A Phase 1b study with 3733 has been initiated and a Phase 1a study with 4334 is planned for the second half of 2022

References

1. Huang Q, et al. *Antimicrob Agents Chemother*. 2020;64:e01463–20.
2. Yuen MF, et al. *Lancet Gastroenterol Hepatol.* 2020;5:152–66.
3. Agarwal K, et al. Poster presentation at: EASL: June 23–26, 2020.
4. Yuen MF, et al. *J Hepatol*. 2022; S0168-8278(22)00238-0.
5. Sulkowski M, et al. *J Hepatol.* 2022; S0168-8278(22)00348-8.
6. Cai D, et al. Oral presentation at EASL: June 23–26, 2021.
7. Unchwaniwala N, et al. Poster presentation at EASL: June 22–26, 2022.
8. Shen M, et al. Poster presentation at EASL: June 22–26, 2022.
9. Xu X, et al. Poster Presentation at AASLD: November 12–25, 2021.

Acknowledgements

- Writing and editorial support was provided by Gregory Suess, PhD, of AlphaBioCom LLC, and funded by Assembly Biosciences, Inc.
- This study was sponsored by Assembly Biosciences, Inc.

assemblybio

Presented at the 2022 International HBV meeting, September 18–22, 2022