Preclinical Profiling of ABI-6250, a Novel Orally Bioavailable Small-Molecule Therapeutic Candidate for the Treatment of Chronic Hepatitis D

WED-377

Marc P Windisch, Nuruddin Unchwaniwala, Jinghu Carl Li, Yanhong Zhu, Heidi Contreras, Dinara Azimova, Francielle Tramontini Gomes de Sousa, Joseph Tan, Kirsten Stray, Ariel Tang, Peter Haggie, Gene Schulze, Michael Shen, Jiaxin Yu, Michel Perron, Michael A Walker, William E Delaney, Min Zhong

Assembly Biosciences, Inc., South San Francisco, CA, USA

Background

- Chronic hepatitis D virus (HDV) infection (cHDV), the most severe form of viral hepatitis, is currently an incurable disease affecting an estimated 12 to 72 million patients worldwide[1,2]
- Patients suffering from cHDV coinfected with hepatitis B virus (HBV) vs HBV alone have an increased risk of developing life-threatening liver conditions, such as fibrosis, cirrhosis, and hepatocellular carcinoma (HCC)[3–5]
 - The incidence rates of HCC are more than 2.5-fold higher in patients with HDV/HBV coinfection vs HBV monoinfection[6]
- Both HBV and the satellite virus HDV use sodium taurocholate cotransporting polypeptide (NTCP), a bile acid transporter on hepatocytes, as an entry receptor into the liver[7–10]
- Bulevirtide (BLV), a peptide inhibitor targeting NTCP and the only drug approved by the European Medicines Agency for cHDV, requires daily injections to prevent HDV and HBV entry into hepatocytes[9,11-14]
- There is a medical need for efficacious, orally administered entry inhibitors that may improve the long-term clinical outcomes of patients suffering from cHDV

Objective

- To describe the preclinical profile of ABI-6250, an orally bioavailable small-molecule entry inhibitor that is a clinical drug candidate for the treatment of cHDV

Methods

- **HDV infection: Measurement of hepatitis D antigen (HDAg):**
 - HepG2-NTCP cells were infected with HDV inoculum and cotreated with ABI-6250. After 24 hours, supernatants were removed and fresh media without ABI-6250 were added. At 5 days postinfection (dpi), HDAg was measured by an in-cell enzyme-linked immunosorbent assay (ELISA) to generate half-maximal effective concentration (EC_{50}) values
- **Human serum shift assay:**
 - HDV-infected cells in the presence of fetal bovine serum (FBS) and physiological levels of human serum albumin (45 mg/mL) and alpha-1-acid glycoprotein (0.7 mg/mL) were compared to a standard infection carried out in media with FBS alone. EC_{50} values were generated by quantifying HDAg at 5 dpi by in-cell ELISA
- **NTCP-mediated bile acid uptake inhibition:**
 - HEK293T cells expressing human NTCP were preincubated for 60 minutes with ABI-6250 or BLV, followed by a 50-minute incubation with 3-α-nitrobenzoxadiazole taurocholic acid. After supernatant removal, fluorescence intensity was measured by plate reader
- **Organic anion-transporting protein (OATP) inhibition:**
 - HEK293T cells expressing human OATP1B1 or OATP1B3 were preincubated for 30 minutes with ABI-6250 or BLV followed by a 10-minute incubation with fluorescein-methotrexate. After supernatant removal, fluorescence intensity was measured by flow cytometry
- **PreS1 binding competition:**
 - HEK293T cells stably expressing human NTCP were coincubated with myristoylated preS1-Alexa-594 peptide and ABI-6250 or BLV for 10 minutes. Binding of the fluorescent peptide was measured by flow cytometry
- **Metabolic stability:**
 - Metabolic stability was determined at 1 μM of testing concentration with non-human primate (NHP) and human liver microsomes (LMs) using liquid chromatography–tandem mass spectrometry (LC-MS)
- **Pharmacokinetic (PK) studies:**
 - ABI-6250 PK parameters were obtained following a single intravenous bolus dose at 1 mg/kg and oral dose at 2.5 or 5 mg/kg in Sprague-Dawley rats and NHPs. Samples were analyzed using LC-MS
- **Pharmacodynamic (PD) studies:**
 - Total bile acid (TBA) levels were measured by enzymatic cycling, and coproporphyrin-I (CP-I) was measured using LC-MS

Results

Figure 1. ABI-6250 Efficiently Inhibited HDV Infection of the Most Prevalent Genotypes

A. Anti-HDV Activity in HepG2-NTCP Cells

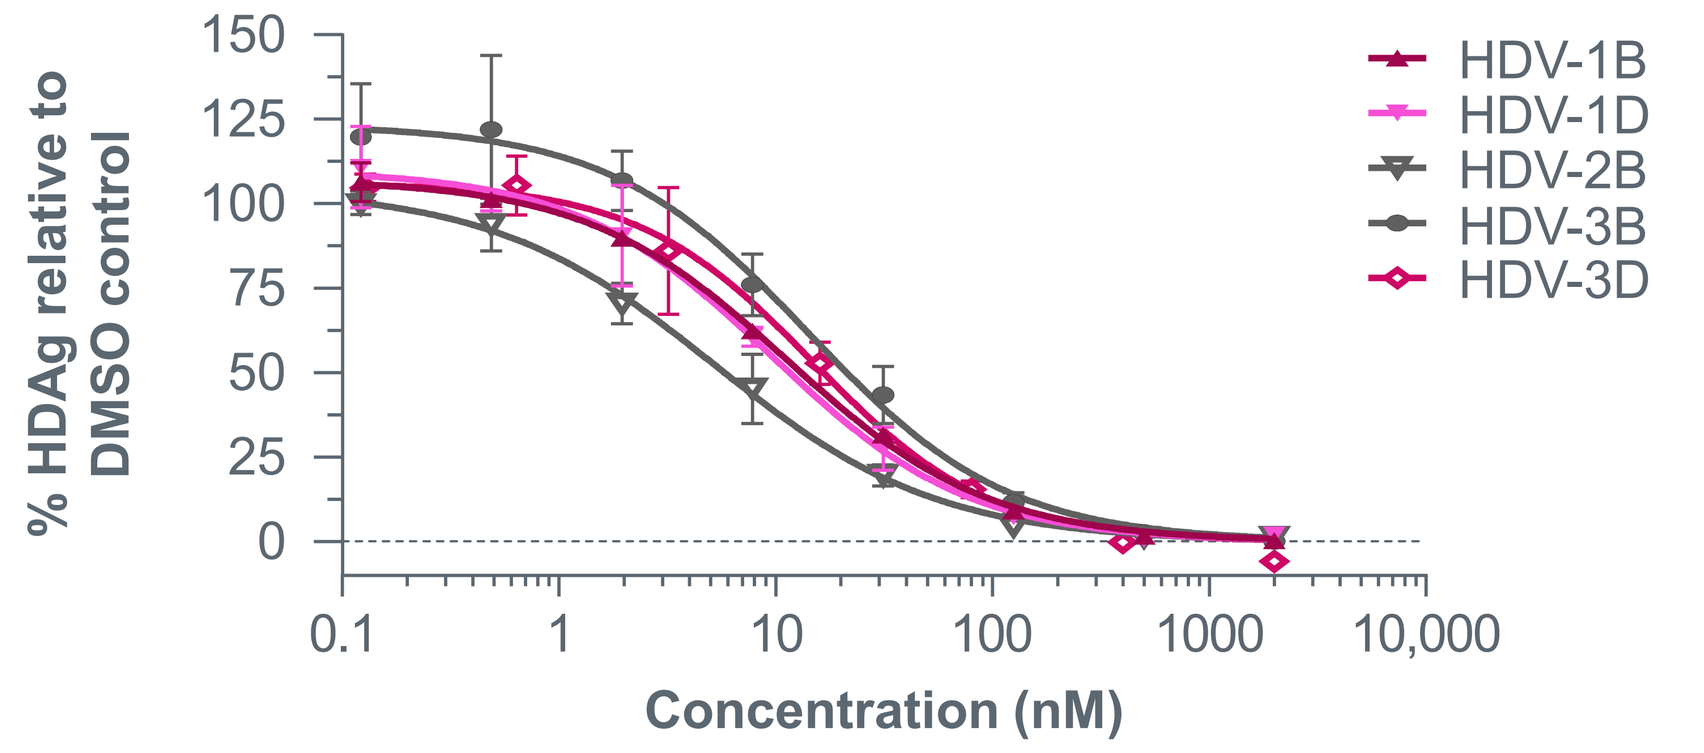
% HDAg relative to DMSO control
150
125
100
75
50
25
0
0.1
1
10
100
1000
10,000
Concentration (nM)
HDV-1B
HDV-1D
HDV-2B
HDV-3B
HDV-3D

B. ABI-6250 EC_{50} Values in HepG2-NTCP Cells

HDV/HBV genotype	EC_{50} (nM)
1B	11.4
1D	9.6
2B	5.2
3B	14.2
3D	14.9

DMSO, dimethyl sulfoxide; EC_{50}, half-maximal effective concentration; HBV, hepatitis B virus; HDAg, hepatitis D antigen; HDV, hepatitis D virus; NTCP, sodium taurocholate cotransporting polypeptide.

- ABI-6250 efficiently inhibited all tested HDV genotypes (1–3) with HBV genotype B or D envelopes (**Figure 1**)
- ABI-6250 efficiently inhibited HDV entry in HepG2-NTCP cells as demonstrated by HDAg in-cell ELISA (**Figure 1A**), with EC_{50} values ranging from 5.2 to 14.9 nM (**Figure 1B**)
- Human serum factors affected the inhibitory potency of ABI-6250 for HDV genotype 3D by increasing the *in vitro* EC_{50} value 35-fold (data not shown)

Figure 2. ABI-6250 Inhibited PreS1-NTCP Binding and NTCP-Mediated Bile Acid Uptake

A. PreS1-NTCP Binding Inhibition

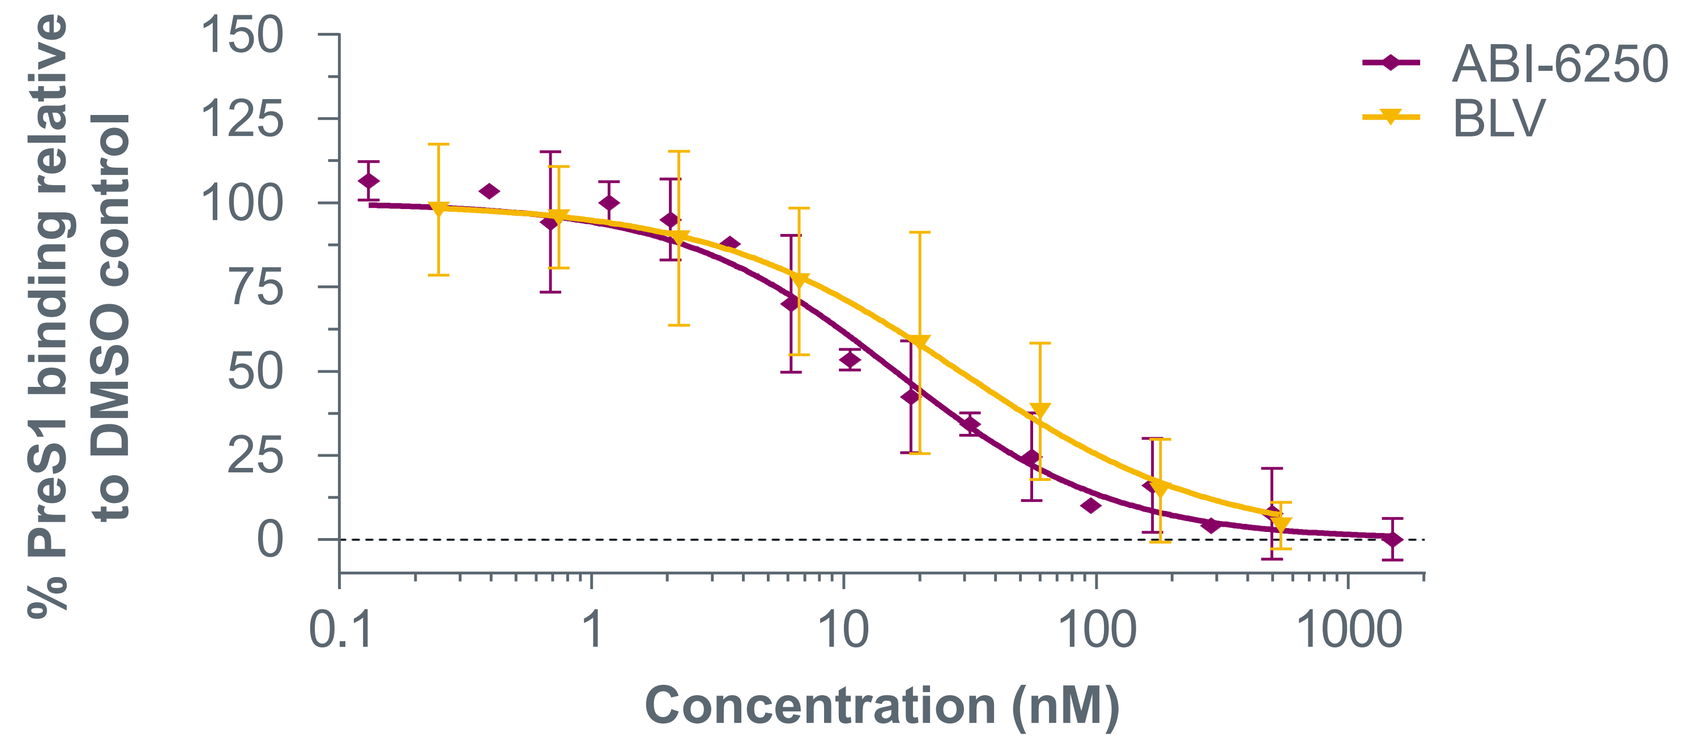
% PreS1 binding relative to DMSO control
150
125
100
75
50
25
0
0.1
1
10
100
1000
Concentration (nM)
ABI-6250
BLV

B. NTCP-Mediated Bile Acid Uptake Inhibition

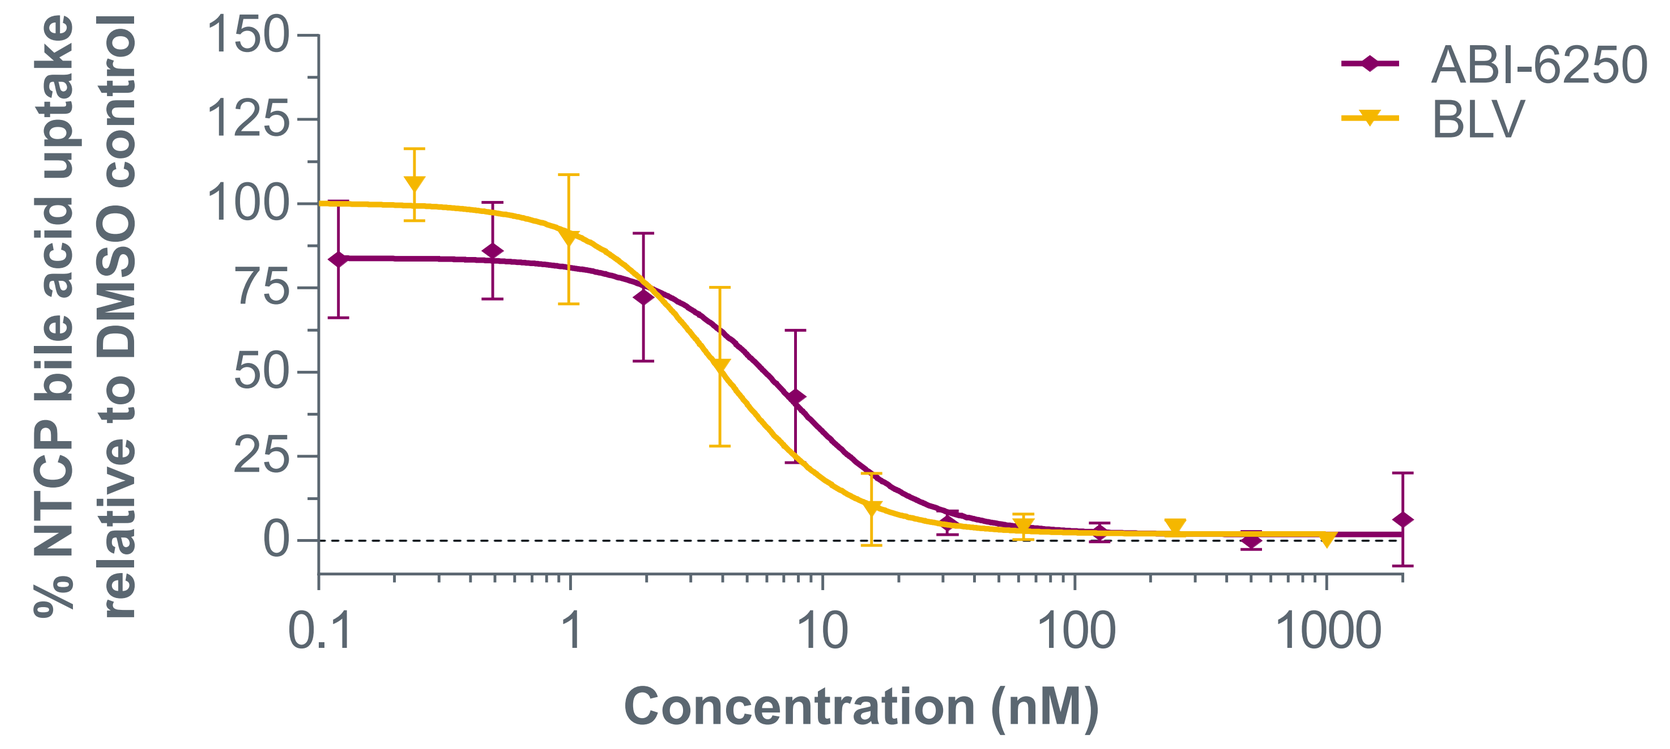
% NTCP bile acid uptake relative to DMSO control
150
125
100
75
50
25
0
0.1
1
10
100
1000
Concentration (nM)
ABI-6250
BLV

BLV, bulevirtide; DMSO, dimethyl sulfoxide; NTCP, sodium taurocholate cotransporting polypeptide.

- ABI-6250 efficiently inhibited HBV preS1-NTCP binding and NTCP-mediated bile acid uptake in HEK293T NTCP cells (**Figure 2**)
- Dose-response curves showed that ABI-6250 and BLV inhibited preS1-NTCP binding, with half-maximal inhibitory concentration (IC_{50}) values of 16.0 and 28.9 nM, respectively (**Figure 2A**)
- Dose-response curves showed that ABI-6250 and BLV inhibited bile acid uptake, with IC_{50} values of 7.3 and 3.9 nM, respectively (**Figure 2B**)

Figure 3. ABI-6250 Selectively Inhibited NTCP

A. NTCP, OATP1B, and ASBT Bile Acid Uptake Inhibition

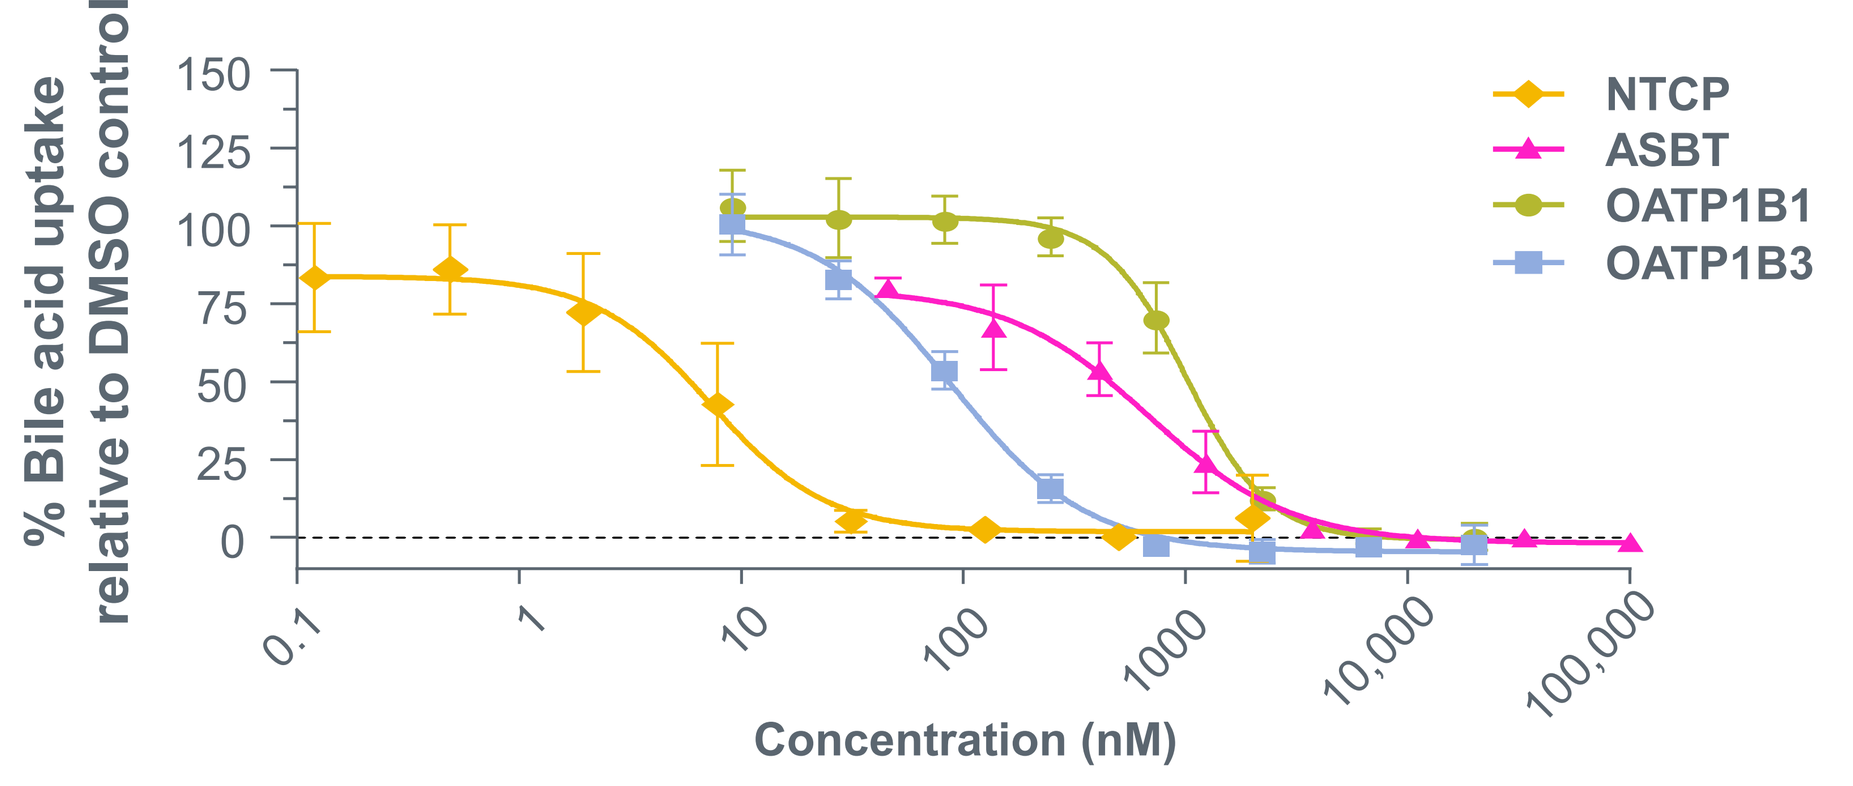
% Bile acid uptake relative to DMSO control
150
125
100
75
50
25
0
0.1
1
10
100
1000
10,000
100,000
Concentration (nM)
NTCP
ASBT
OATP1B1
OATP1B3

B. ABI-6250 Fold Selectivity

Transporter	ABI-6250 IC_{50} (nM) [fold selectivity][a]
NTCP	7.3
OATP1B1	1000 [>135]
OATP1B3	90 [>12]
ASBT	700 [>95]

[a]Numbers in brackets indicate fold selectivity calculated as the ratio of IC_{50} bile acid transporter to IC_{50} NTCP.
ASBT, apical sodium-dependent bile acid transporter; DMSO, dimethyl sulfoxide; IC_{50}, half-maximal inhibitory concentration; OATP, organic anion-transporting protein; NTCP, sodium taurocholate cotransporting polypeptide.

- ABI-6250 had limited inhibitory effects *in vitro* on OATP1B, NTCP, and apical sodium-dependent bile acid transporter (ASBT) activity (**Figure 3**)
- ABI-6250 inhibited OATP1B1-, OATP1B3-, and ASBT-dependent bile acid uptake, with IC_{50} values of 1000, 90, and 700 nM, respectively (**Figure 3A and B**)
- ABI-6250 is a selective NTCP inhibitor, as demonstrated by fold selectivity (calculated as the ratio of IC_{50} bile acid transporter to IC_{50} NTCP; **Figure 3B**)

Figure 4. ABI-6250 Has Favorable ADME and PK/PD Profiles

A. ABI-6250 ADME and PK Profile[a]

ADME properties		
Percentage of ABI-6250 remaining at 45 min after incubation in LMs (human \| NHP \| rat \| mouse)	97 \| 97 \| 97 \| 93	
Caco-2 A-B[b] \| B-A[b] \| ER	4.8 \| 4.7 \| 1.0	
PK properties	**Rat**	**NHP**
$t_{1/2}$ (hr)	4.9[c] \| 5.4[d]	6.7[c] \| 14.8[d]
%F	100	99
CL (mL/min/kg)	18.9	0.6
Vss (L/kg)	4.4	0.6

B. TBA and CP-I Measurements[e]

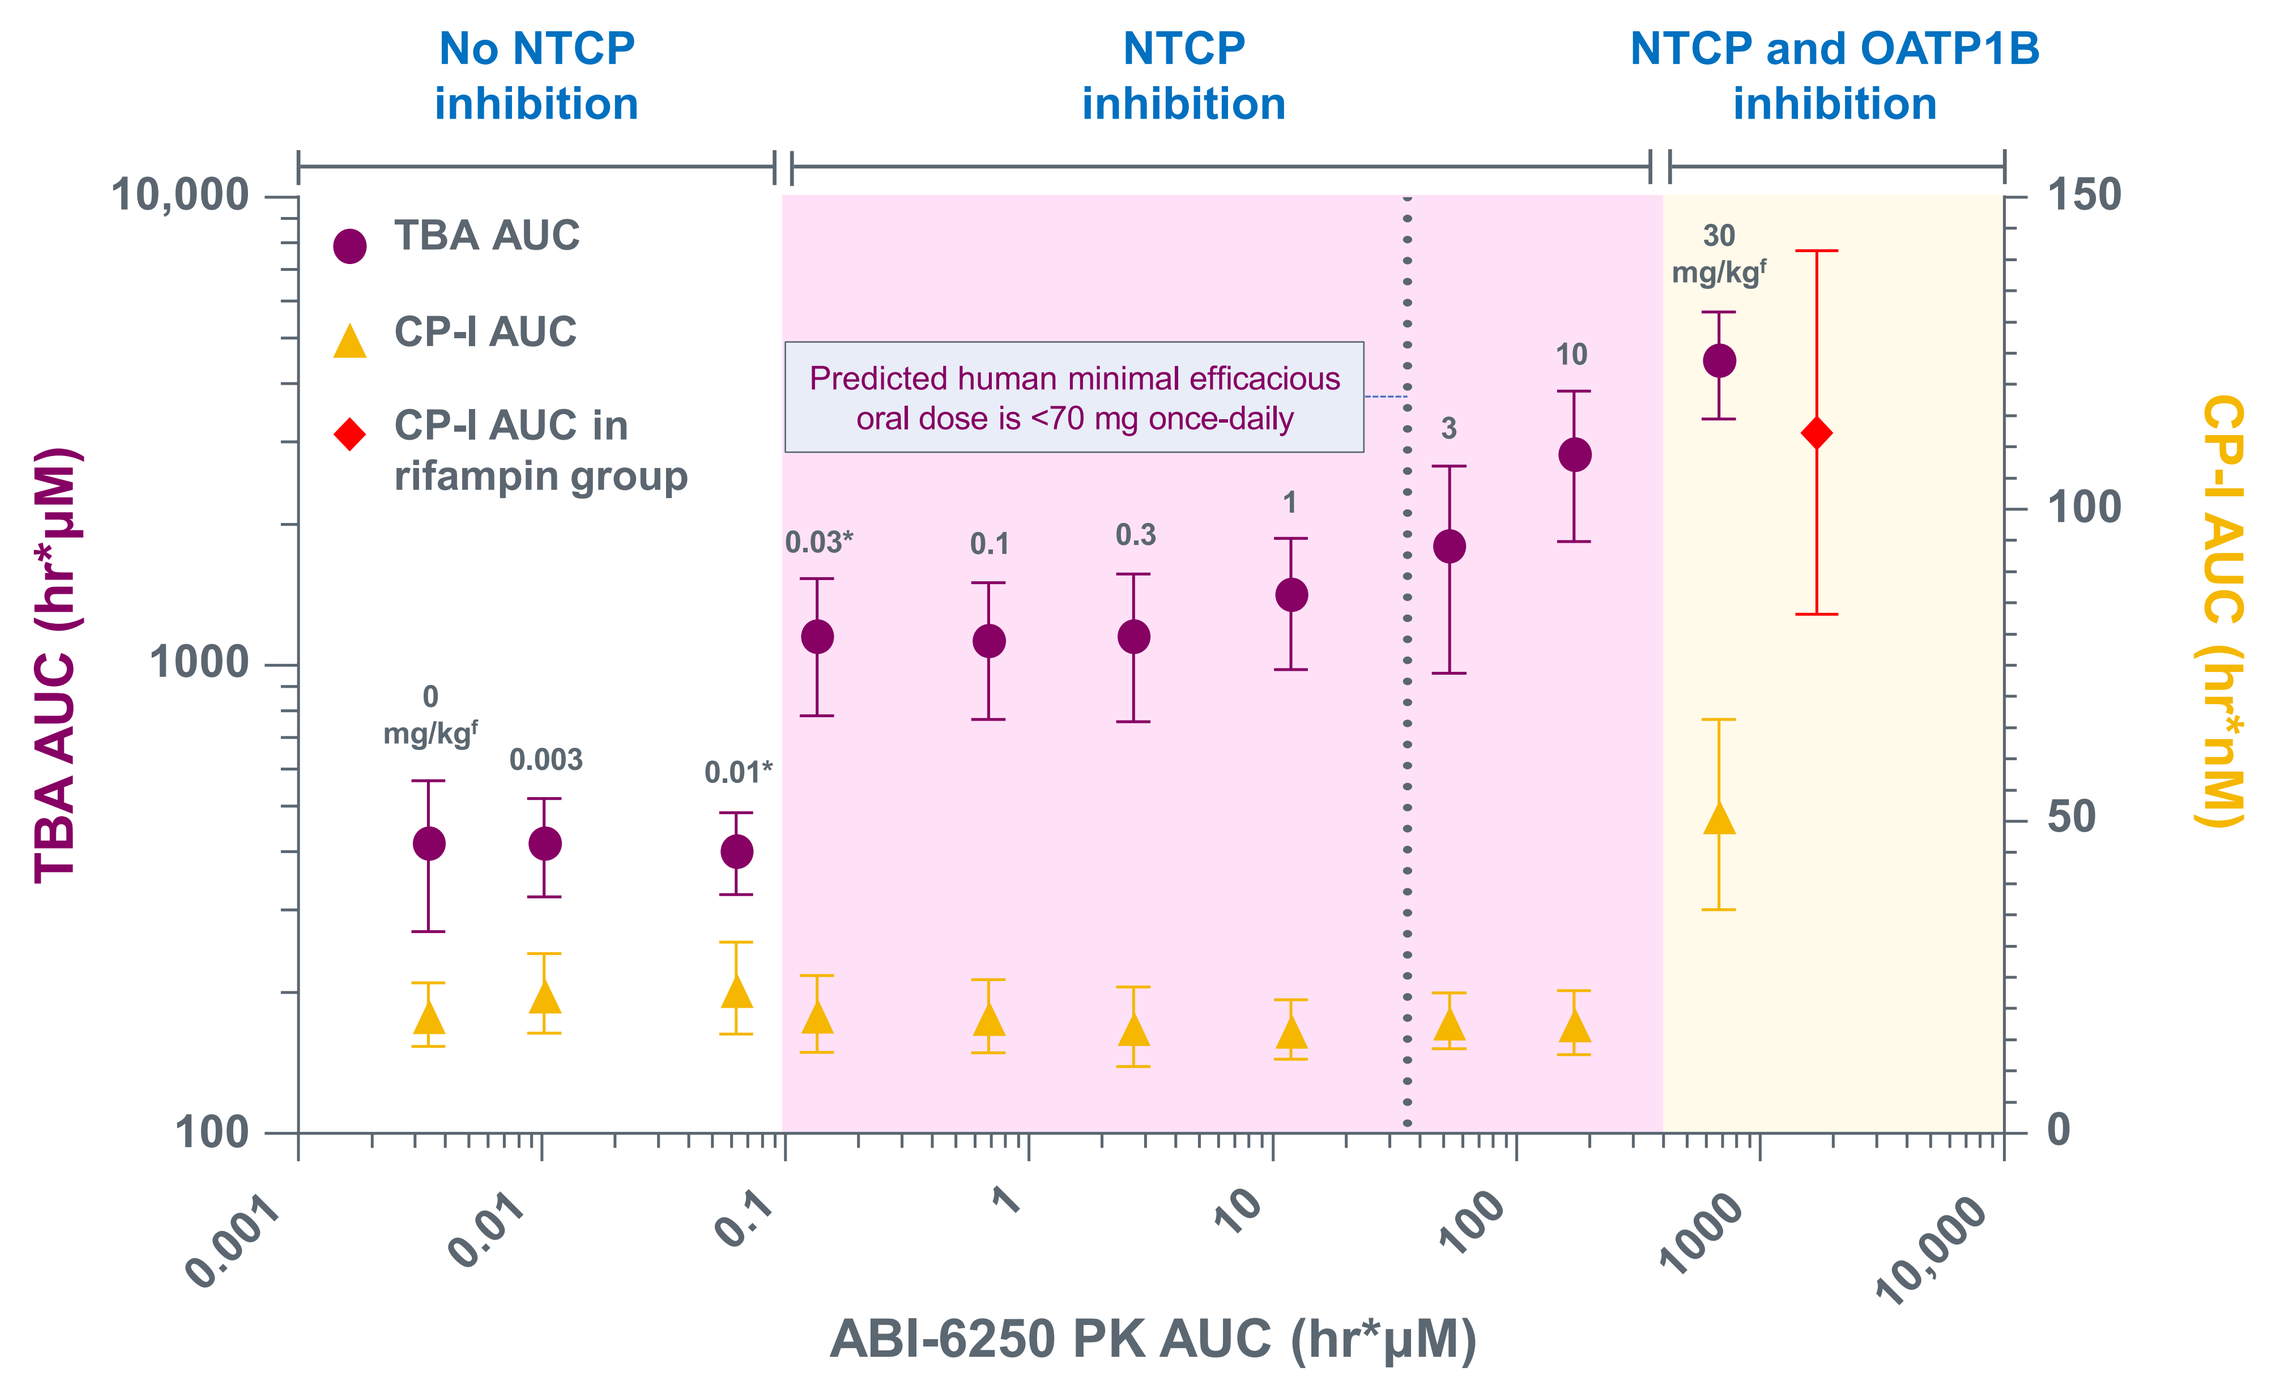
No NTCP inhibition
NTCP inhibition
NTCP and OATP1B inhibition
TBA AUC (hr*μM)
CP-I AUC (hr*nM)
10,000
1000
100
150
100
50
0
TBA AUC
CP-I AUC
CP-I AUC in rifampin group
Predicted human minimal efficacious oral dose is <70 mg once-daily
0 mg/kg[f]
0.003
0.01*
0.03*
0.1
0.3
1
3
10
30 mg/kg[f]
0.001
0.01
0.1
1
10
100
1000
10,000
ABI-6250 PK AUC (hr*μM)

[a]n = 2 or 3. [b]10^{-6} cm/s. [c]If ABI-6250 is administered intravenously. [d]If ABI-6250 is administered orally. [e]The x-axis values for vehicle, 0.003 mg/kg and rifampin groups are arbitrarily set. [f]Doses of ABI-6250 in NHPs; n = 6 per dose. *For TBA AUC P <0.01 between 0.01 mg/kg and 0.03 mg/kg. Vertical line indicates human PK projection using allometric scaling.
A-B, A to B permeability; ADME, absorption, distribution, metabolism, and excretion; AUC, area under the curve; B-A, B to A permeability; CL, clearance; CP-I, coproporphyrin-I; ER, efflux ratio; %F, bioavailability; LM, liver microsome; NHP, non-human primate; PD, pharmacodynamics; PK, pharmacokinetics; $t_{1/2}$, terminal half-life; TBA, total bile acid; Vss, volume of distribution.

- Absorption, distribution, metabolism, and excretion (ADME) and PK/PD studies showed that ABI-6250 has:
 - A favorable ADME and PK/PD profile (**Figure 4**)
 - Good LM stability in human and preclinical species (**Figure 4A**)
 - Good apparent permeability in Caco-2 cells, with no efflux and terminal half-lives ($t_{1/2}$) in rat and NHPs greater than 4.9 hours. The predicted $t_{1/2}$ in humans is over 30 hours when administered orally
 - Approximately 100% bioavailability in NHPs and rats, with $t_{1/2}$ ranging from 4.6 to 15.7 hours
 - Elevated TBA levels at doses up to 30 mg/kg in NHPs, indicating drug-target engagement. CP-I levels, a biomarker for OATP1B inhibition, were not increased at physiologic concentrations of ABI-6250. Rifampin, an OATP1B inhibitor, efficiently elevated CP-I levels (**Figure 4B**)
 - No cytochrome P450 inhibition and human ether-a-go-go-related gene liabilities (data not shown)
 - Demonstrated good metabolic stability, with more than 95% remaining after 45 minutes of incubation in NHP and human LMs (**Figure 4A**)
 - The potential to achieve the desired minimum efficacious concentration coverage with <70 mg once-daily dosing

Conclusions

- ABI-6250 is an NTCP-selective, highly potent, orally bioavailable HDV entry inhibitor
- At relevant concentrations, ABI-6250 elevates total bile acids in non-human primates, indicating target engagement without increasing coproporphyrin-I plasma levels, a biomarker for OATP1B inhibition
- The preclinical PK profile of ABI-6250 supports low once-daily dosing in patients with cHDV
- ABI-6250 is expected to enter Phase 1 clinical trials by the end of 2024

REFERENCES

1) World Health Organization. Hepatitis D Fact Sheet. https://www.who.int/news-room/fact-sheets/detail/hepatitis-d. Accessed April 4, 2024. **2)** Negro F, et al. *JAMA*. 2023;330(24):2376-87. **3)** Bockman J, et al. *BMC Gastroenterol*. 2020;20(1):24. **4)** Kamal H, et al. *Hepatology*. 2020;72(4):1177-90. **5)** Manesis E, et al. *J Hepatol.* 2013;59(5):949-56. **6)** Elsaid M, et al. Poster presented at: AASLD 2023, The Liver Meeting; November 10–14, 2023; Boston, MA. **7)** Hughes S, et al. *Lancet*. 2011;278(9785):73-85. **8)** Urban S, et al. *Gut*. 2021;70(9):1782-94. **9)** Wedemeyer H, et al. *N Engl J Med.* 2023;389(1):22-32. **10)** Yan H, et al. *eLife.* 2012;1:300049. **11)** Ni Y, et al. *Gastroenterology.* 2014;146(4):1070-83. **12)** Urban S, et al. *Gastroenterology*. 2014;147(1):48-64. **13)** Kang C, et al. *Drugs*. 2020;80(15):1601-5. **14)** European Medicines Agency. Hepcludex. https://www.ema.europa.eu/en/medicines/human/EPAR/hepcludex. Accessed April 4, 2024.

ACKNOWLEDGEMENTS

- Writing and editorial support were provided by Gregory Suess, PhD, CMPP, of Red Nucleus and were funded by Assembly Biosciences, Inc.
- This study was sponsored by Assembly Biosciences, Inc.
- Contact: Mwindisch@assemblybio.com

DISCLOSURES

- All authors are employees of Assembly Biosciences, Inc., and may own stock

Presented at the European Association for the Study of the Liver (EASL) – The International Liver Congress 2024, June 5–8, 2024